DESIGN AND SYNTHESIS OF RETIFEROLS AS A NEW CLASS OF *DES* - CD VITAMIN D ANALOGS

Part-1: Synthesis of intermediates, optimisation of coupling conditions and evaluation for VDR binding affinity

N. Rao Bolla[1], Aoife Corcoran[2], M. Chodynski[1], E. Marcinkowska[2], G. Brown[3], A. Kutner[1]

[1]Pharmaceutical Research Institute, 01-793 Warsaw, Poland; [2]Faculty of Biotechnology, University of Wroclaw, 50-383 Wroclaw, Poland;
[3]School of Immunity and Infection, University of Birmningham, B15 2TT, UK

I. Introduction:

- Retiferols-Conceptually a novel class of des-CD vitamin D analogs
- Prof. A Kutner (1995) first attempted to simplify the structure and to define the structural parts of vitamin D truly responsible for its activity, des-CD analogs conceived

Rationale for retiferols:

- Basically CD ring part is derived from cholesterol, not synthesized specifically for vitamin D like the triene part or hydroxyl functions
- The CD fragment doesn't participate in activating or deactivating metabolism of vitamin D
- Molecular modeling studies showed that replacement of CD-ring with an aliphatic or olefi nic spacer sufficiently preserves the 3D arrangement of C-1, C-3 and C-25 hydroxyls which are involved in receptor binding
- Modeling also revealed the structural correlation between des-CD vitamins and 4-hydroxy and 2,3 dioxygenated metabolites of retinoids
- All these, supported the designing of des-CD analogs and introduction of new term retiferols

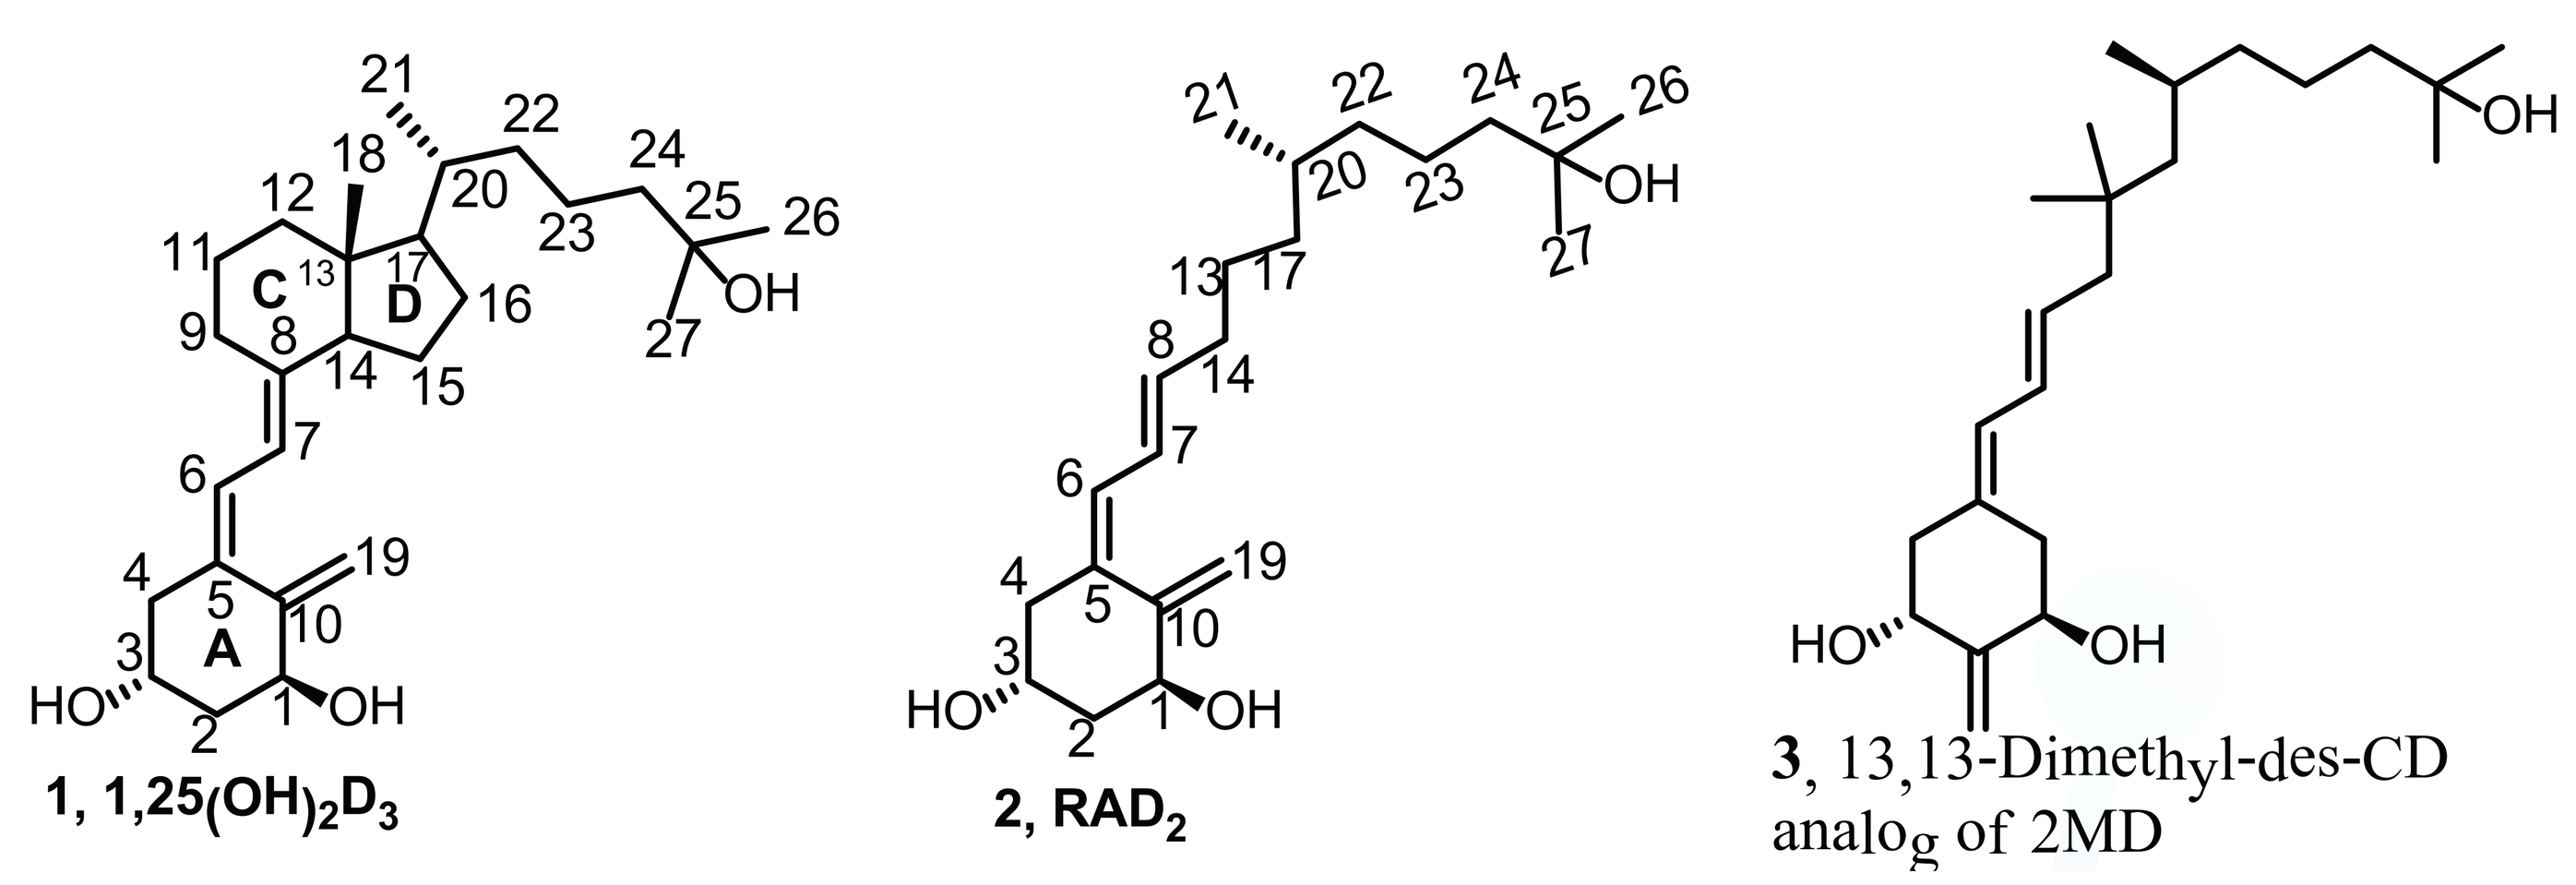

1, 1,25(OH)2D3
2, RAD2
3, 13,13-Dimethyl-des-CD analog of 2MD

Figure 1: Structures fo 1,25-$(OH)_2D_3$ and retiferols RAD_2 & 13,13-dimethyl-*des*-CD-2MD

II. Objectives:

- Designing new A-ring fragments
- Synthesis of designed A ring fragments
- Designing new aliphatic side chain fragments
- Synthesis of designed aliphatic chain frag ments
- Optimisation of conditions for the coupling of A-ring and aliphatic side chain
- Coupling of both A-ring and aliphatic chain fragments to synthesize new retiferols

III. Structures of designed retiferols:

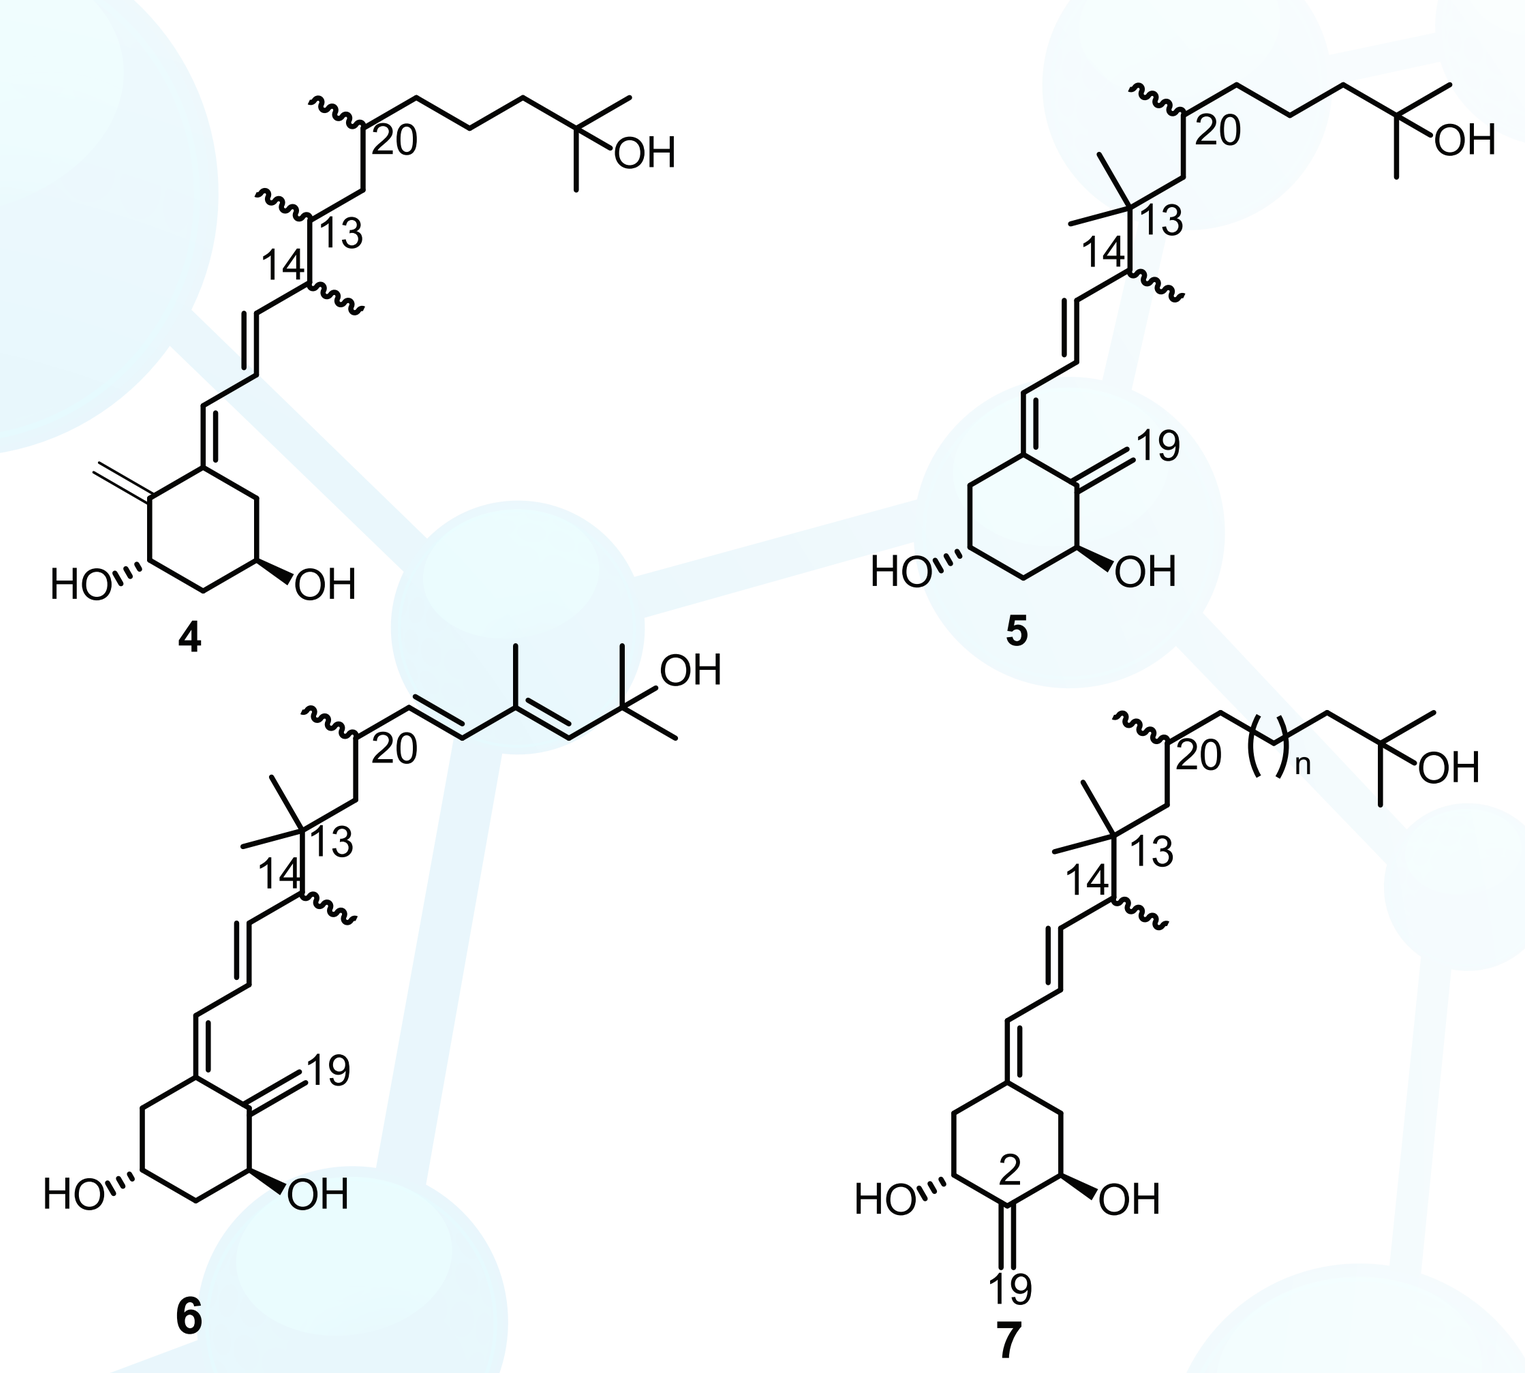

IV. Synthesis of A-ring fragments:

1.Synthesis of A-ring fragment (13):

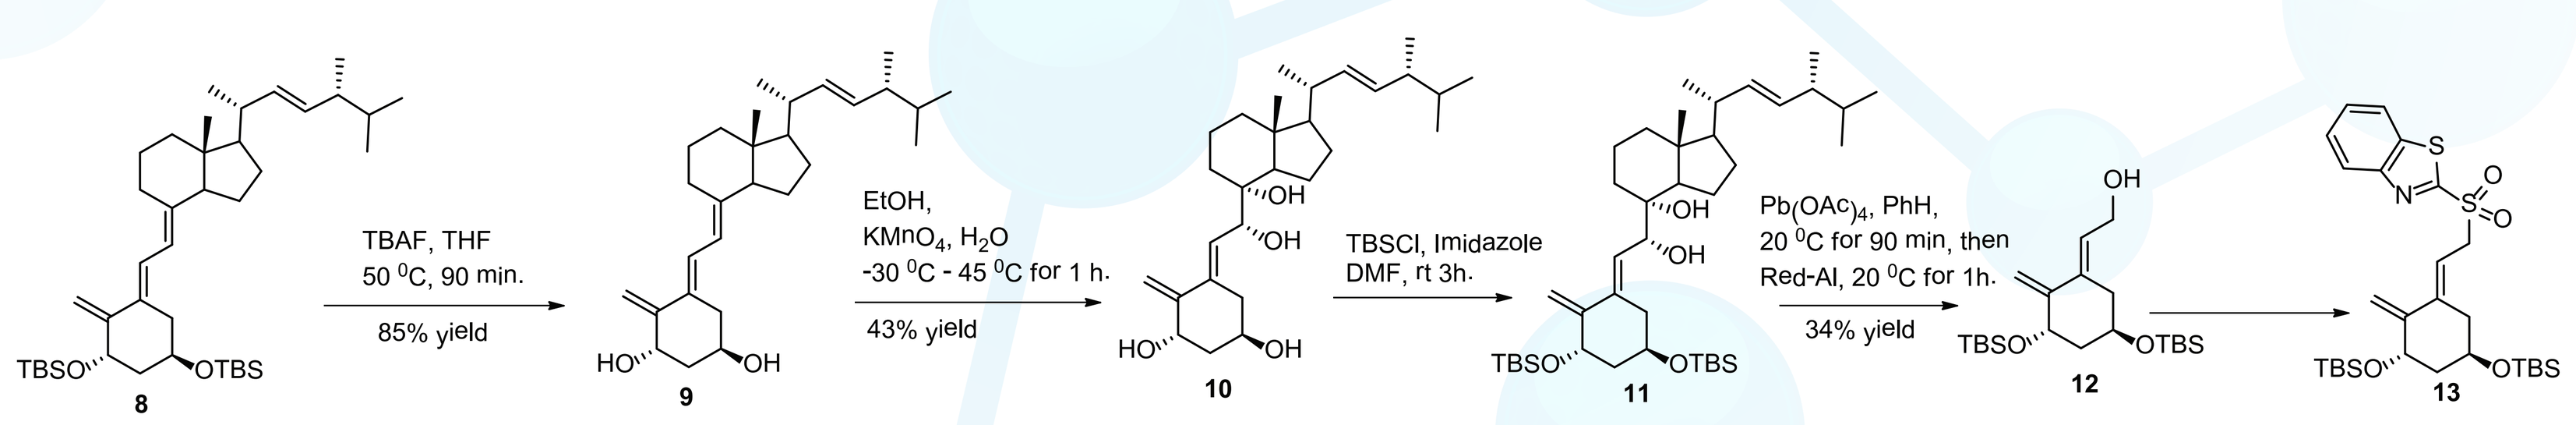

TBAF, THF
50 °C, 90 min.
85% yield
EtOH, KMnO4, H2O
-30 °C - 45 °C for 1 h.
43% yield
TBSCl, Imidazole
DMF, rt 3h.
Pb(OAc)4, PhH,
20 °C for 90 min, then
Red-Al, 20 °C for 1h.
34% yield

2.Synthesis of A-ring fragment (19):

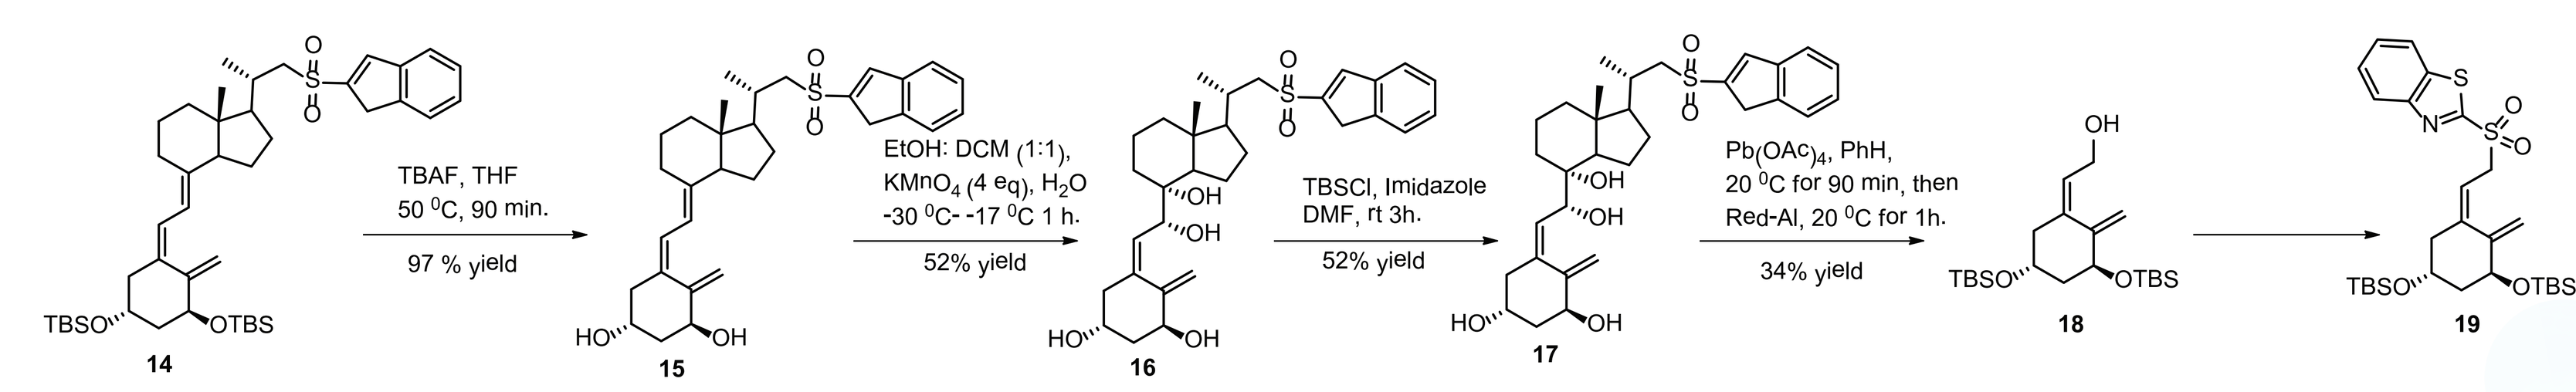

TBAF, THF
50 °C, 90 min.
97 % yield
EtOH: DCM (1:1),
KMnO4 (4 eq), H2O
-30 °C- -17 °C 1 h.
52% yield
TBSCl, Imidazole
DMF, rt 3h.
52% yield
Pb(OAc)4, PhH,
20 °C for 90 min, then
Red-Al, 20 °C for 1h.
34% yield

V. Optimisation of coupling conditions:

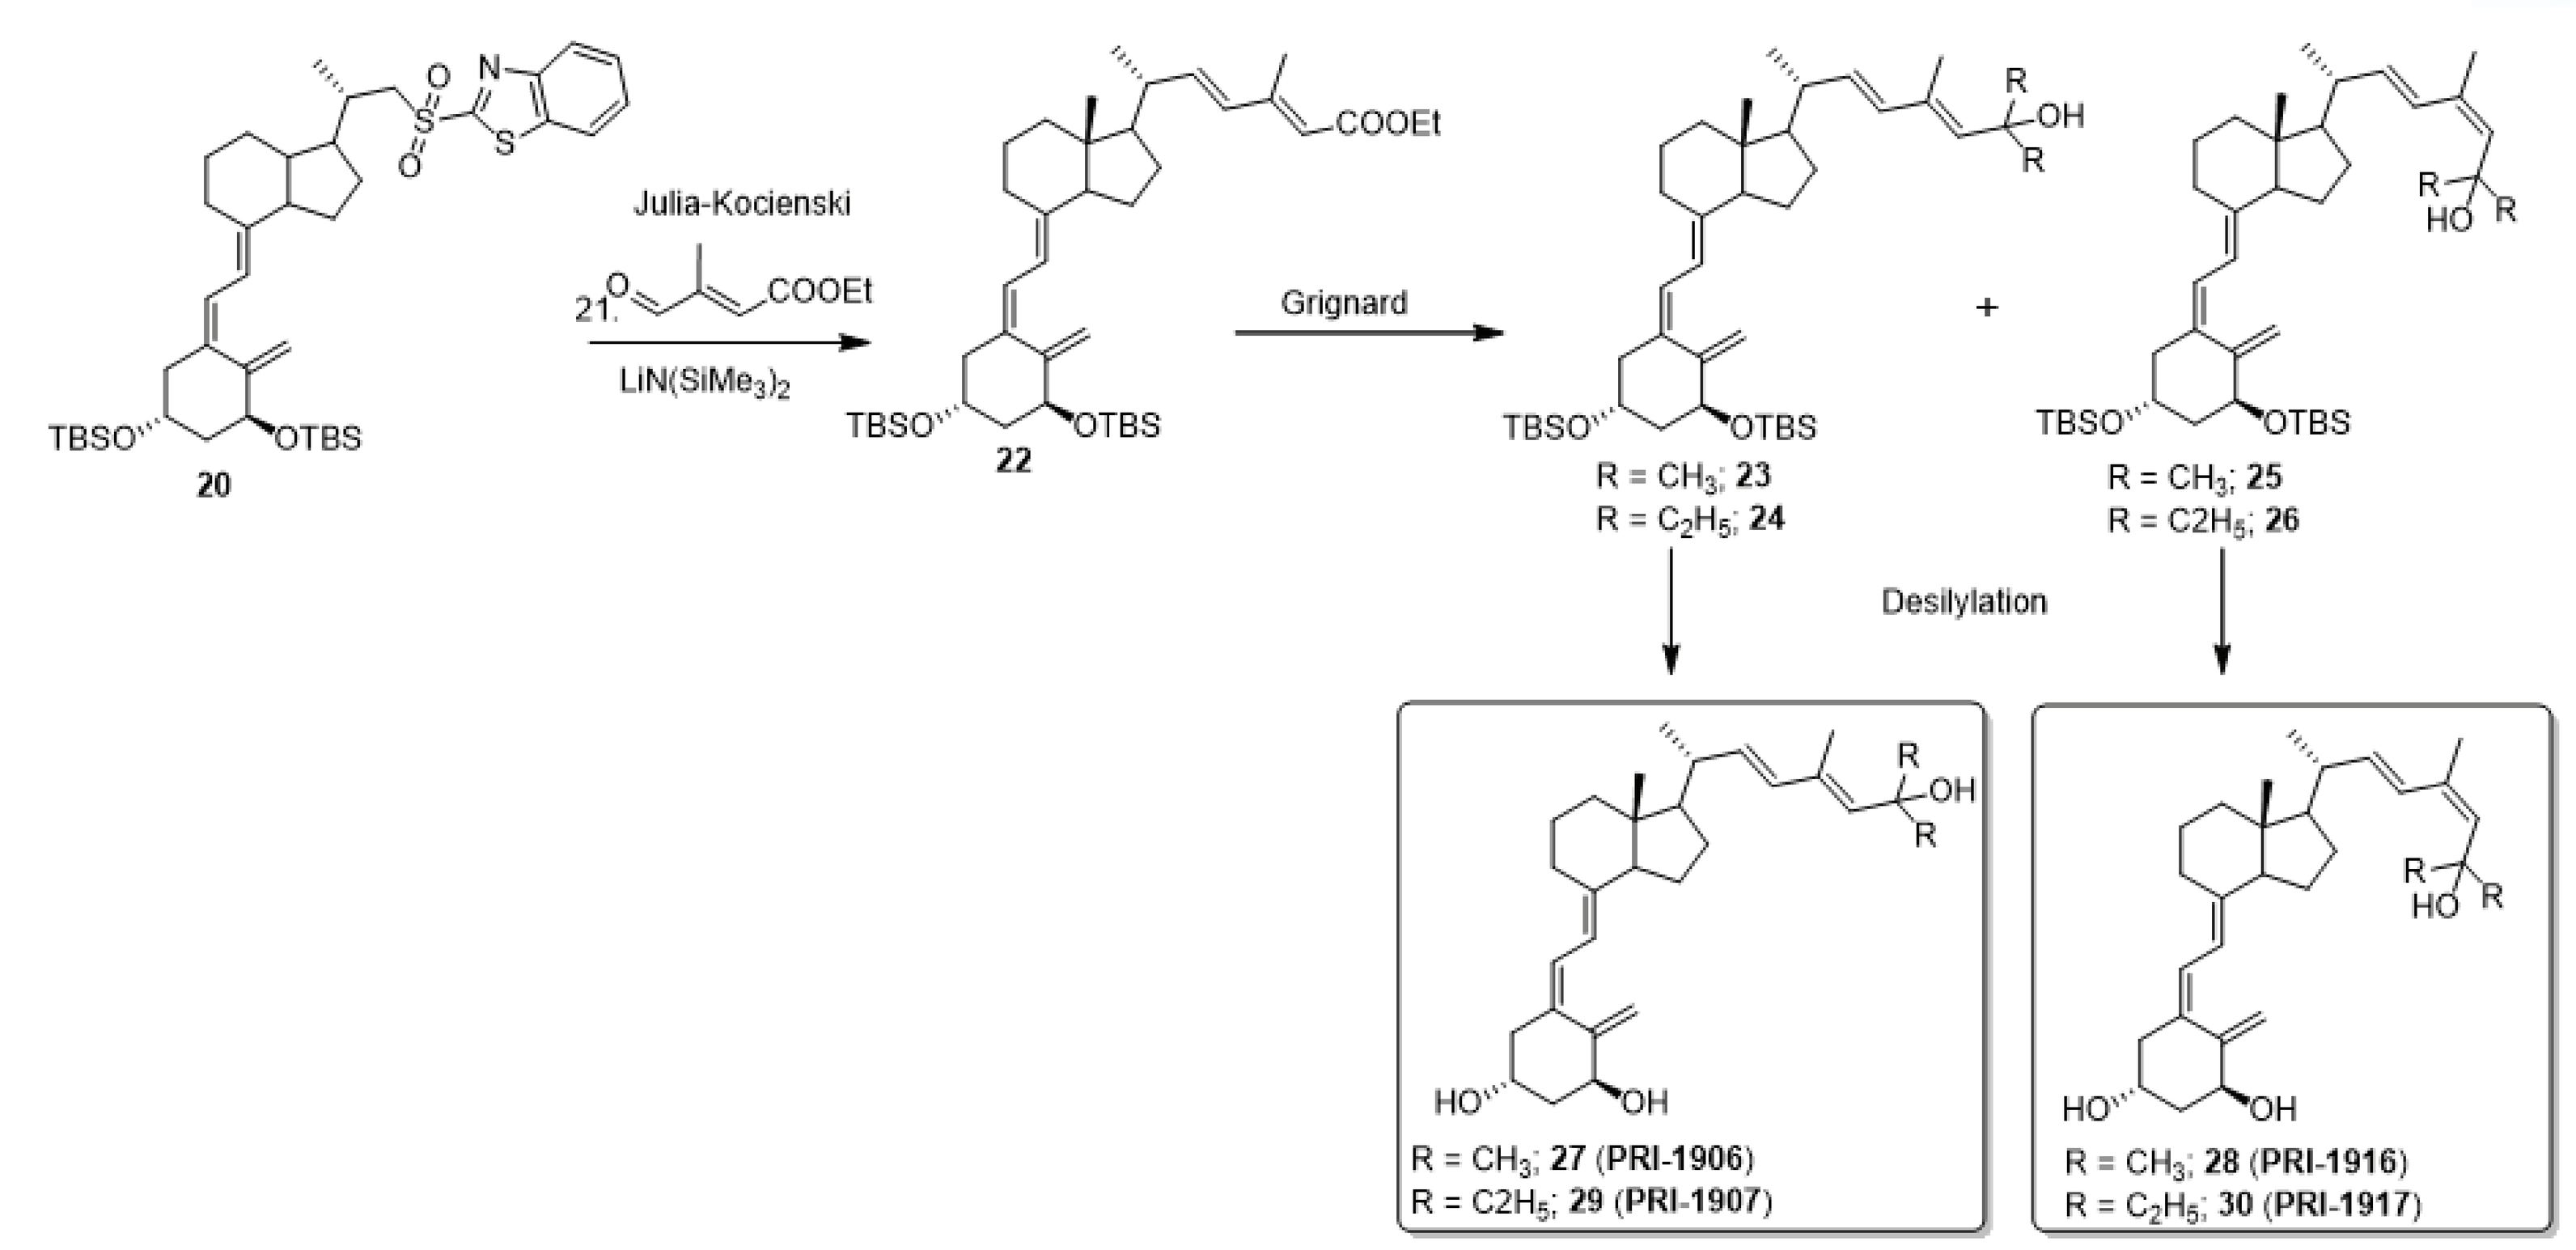

Julia-Kocienski
LiN(SiMe3)2
Grignard
R = CH3; 23
R = C2H5; 24
R = CH3; 25
R = C2H5; 26
Desilylation
R = CH3; 27 (PRI-1906)
R = C2H5; 29 (PRI-1907)
R = CH3; 28 (PRI-1916)
R = C2H5; 30 (PRI-1917)

Figure 4: Scheme for the synthesis of **PRI-1906** and **PRI-1907** and their 24a diastereoisomers **PRI-1916** and **PRI-1917**

VI. Hypothetical structures of 22Z-analogs:

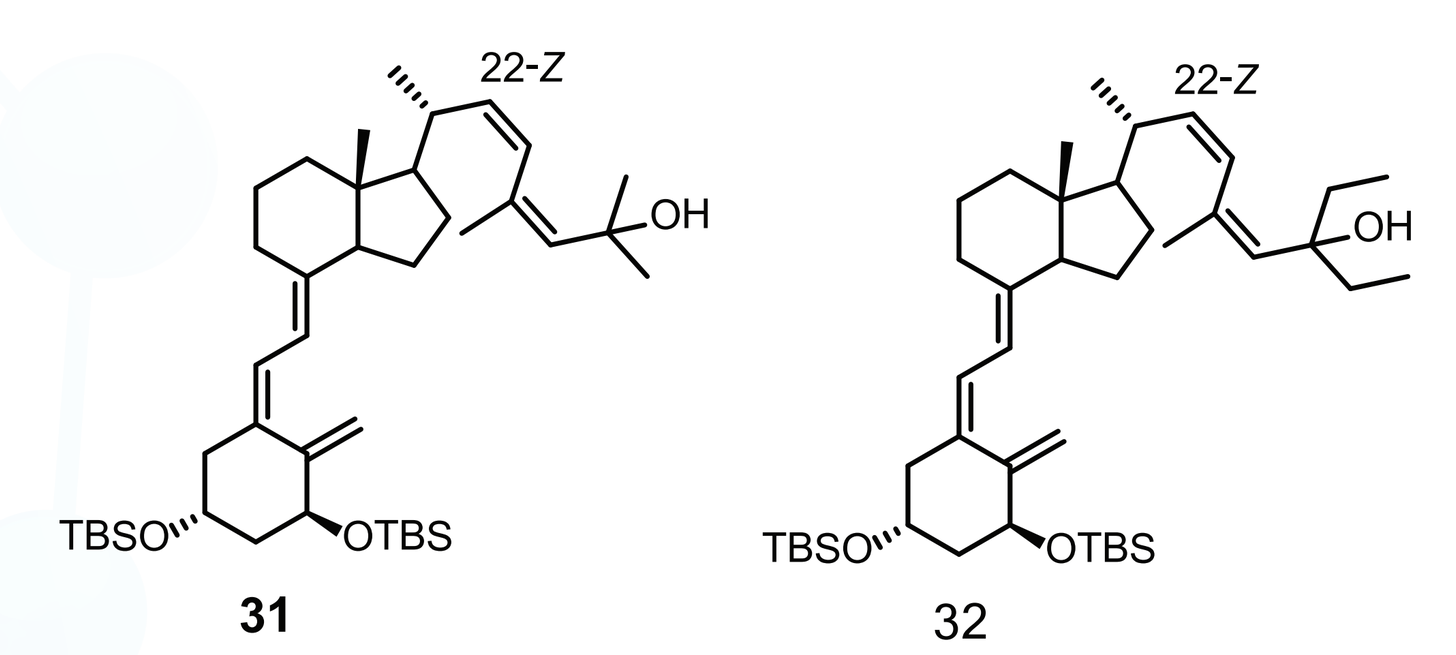

VII. NOE NMR experimental proof for the new structures:

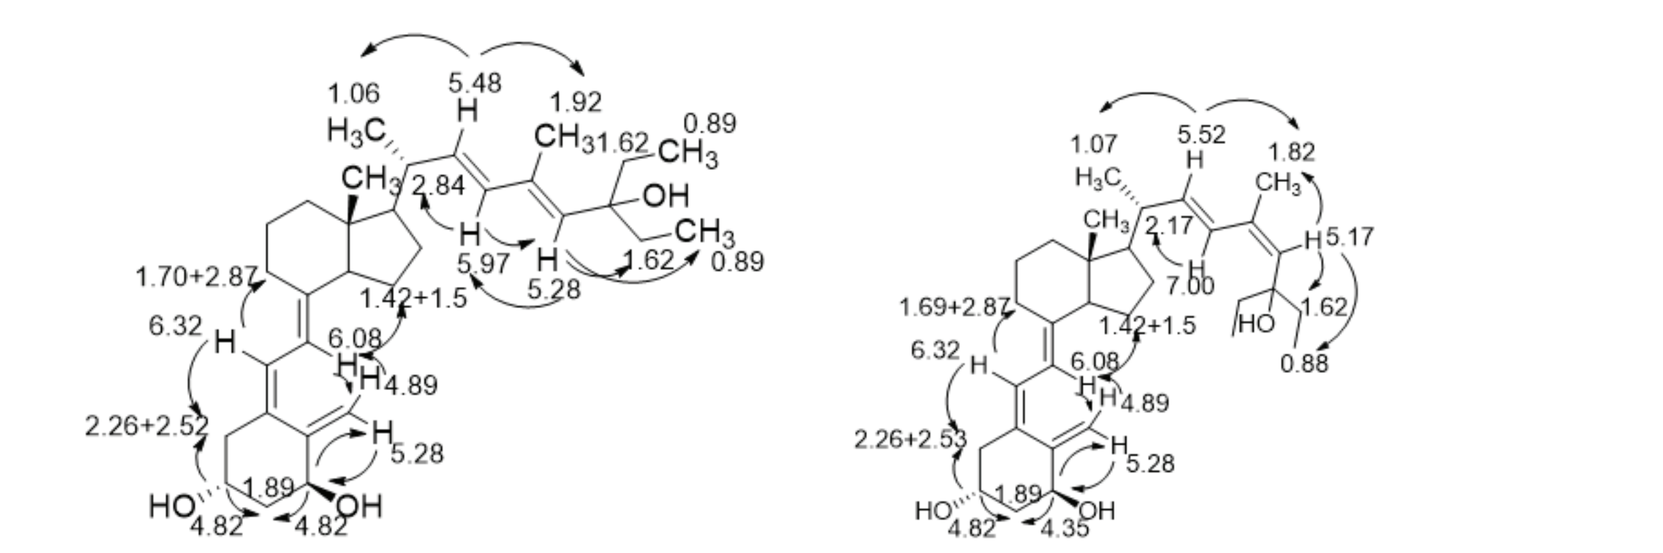

Figure 2: Structure of **29** (**PRI-1907**) showing NOE assignments for characteristic protons

Figure 3: Structure of **30** (**PRI-1917**) showing NOE assignments for characteristic protons

VIII. 600 MHz-1H NMR Spectra showing NOE effects:

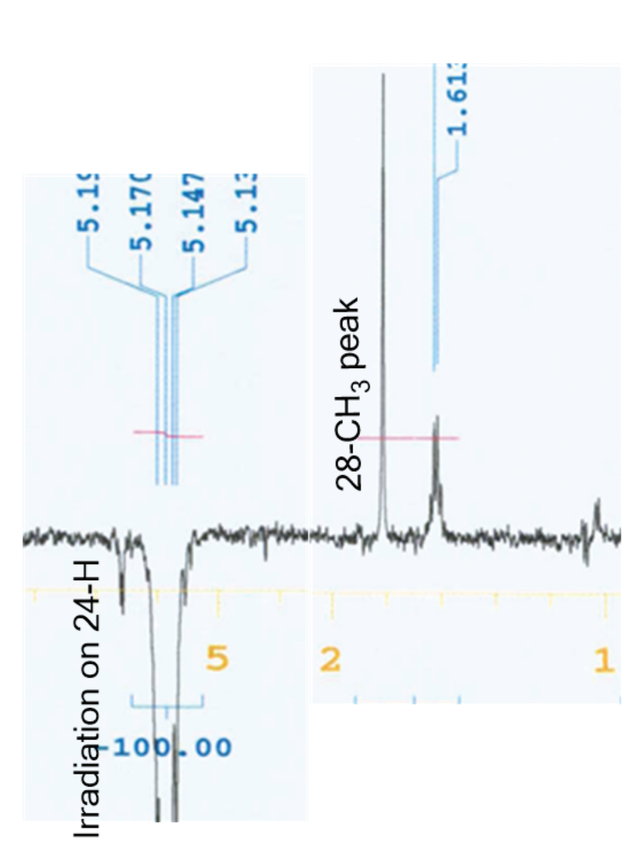

NOE spectrum for compound PRI-1907

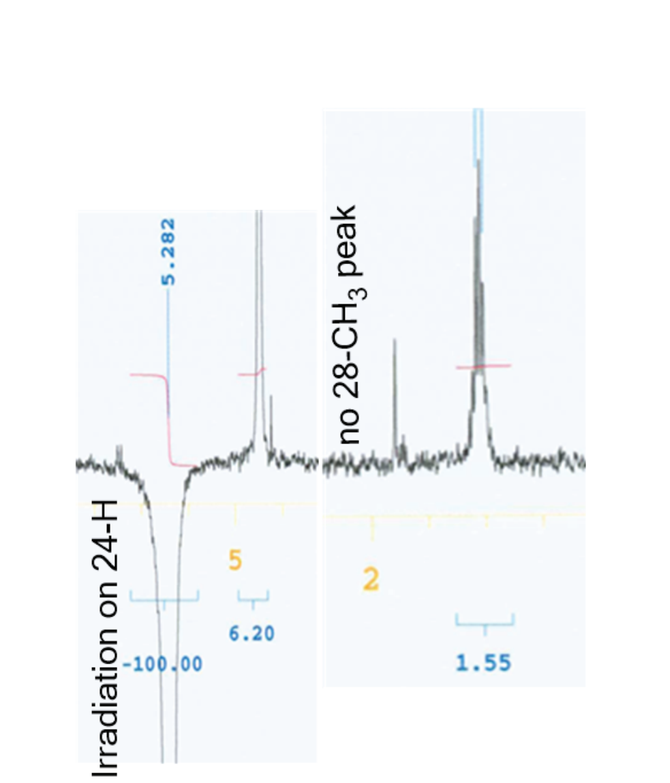

NOE spectrum for compound PRI-1917

IX. 600 MHz-NMR spectra of PRI-1916 and PRI-1917

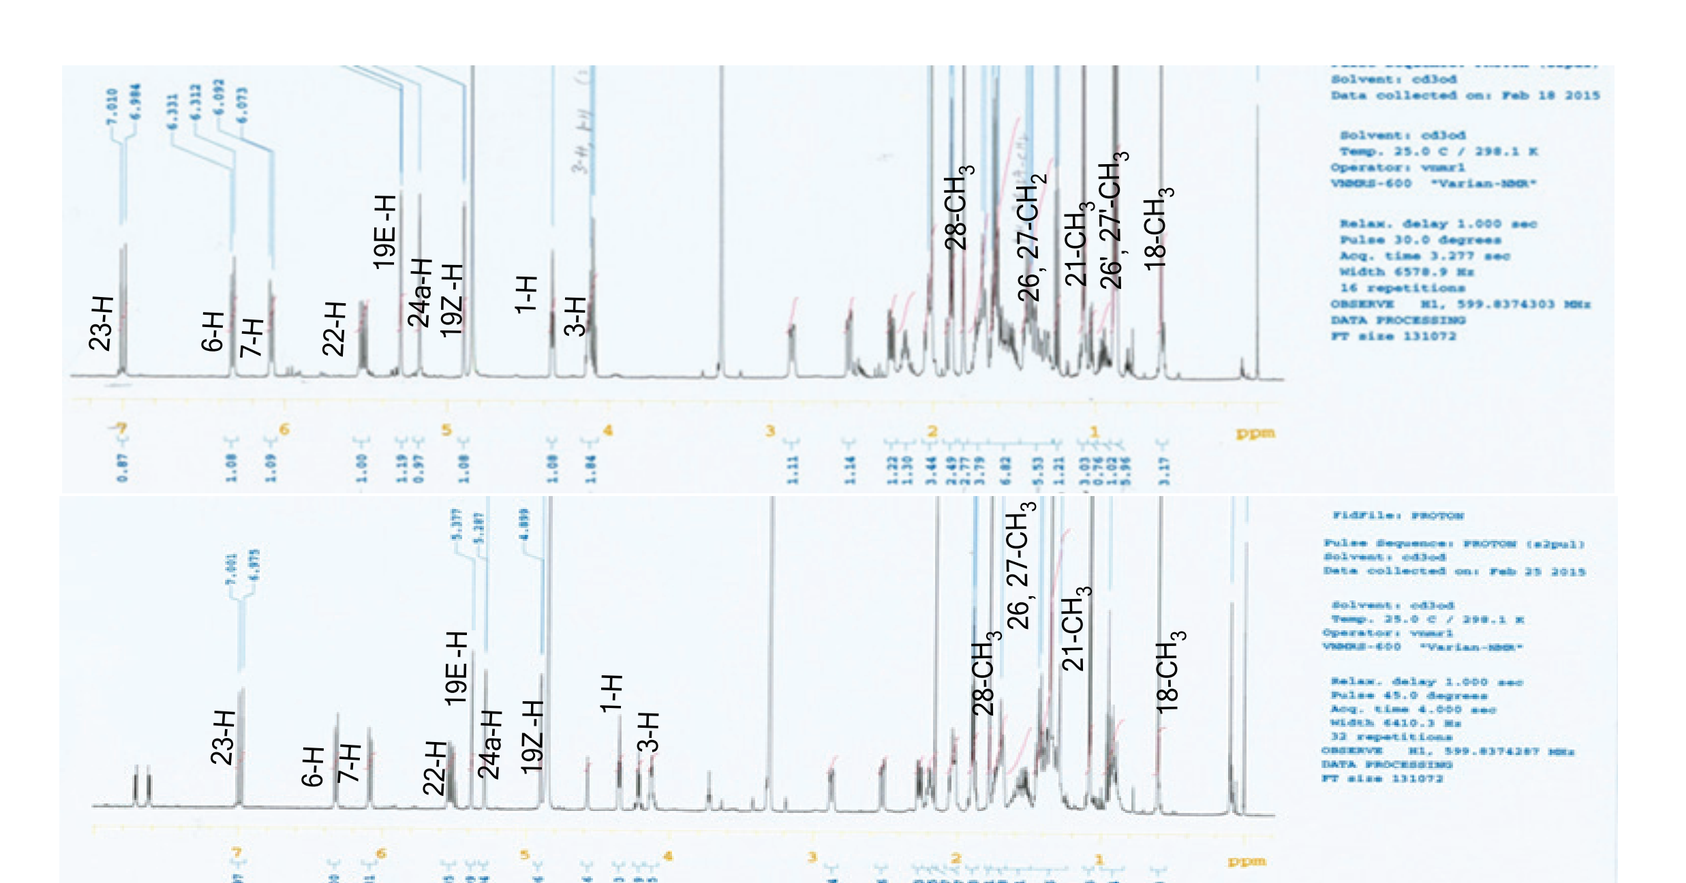

X. 600 MHz ^{13}C and DEPT NMR spectra of PRI-1917

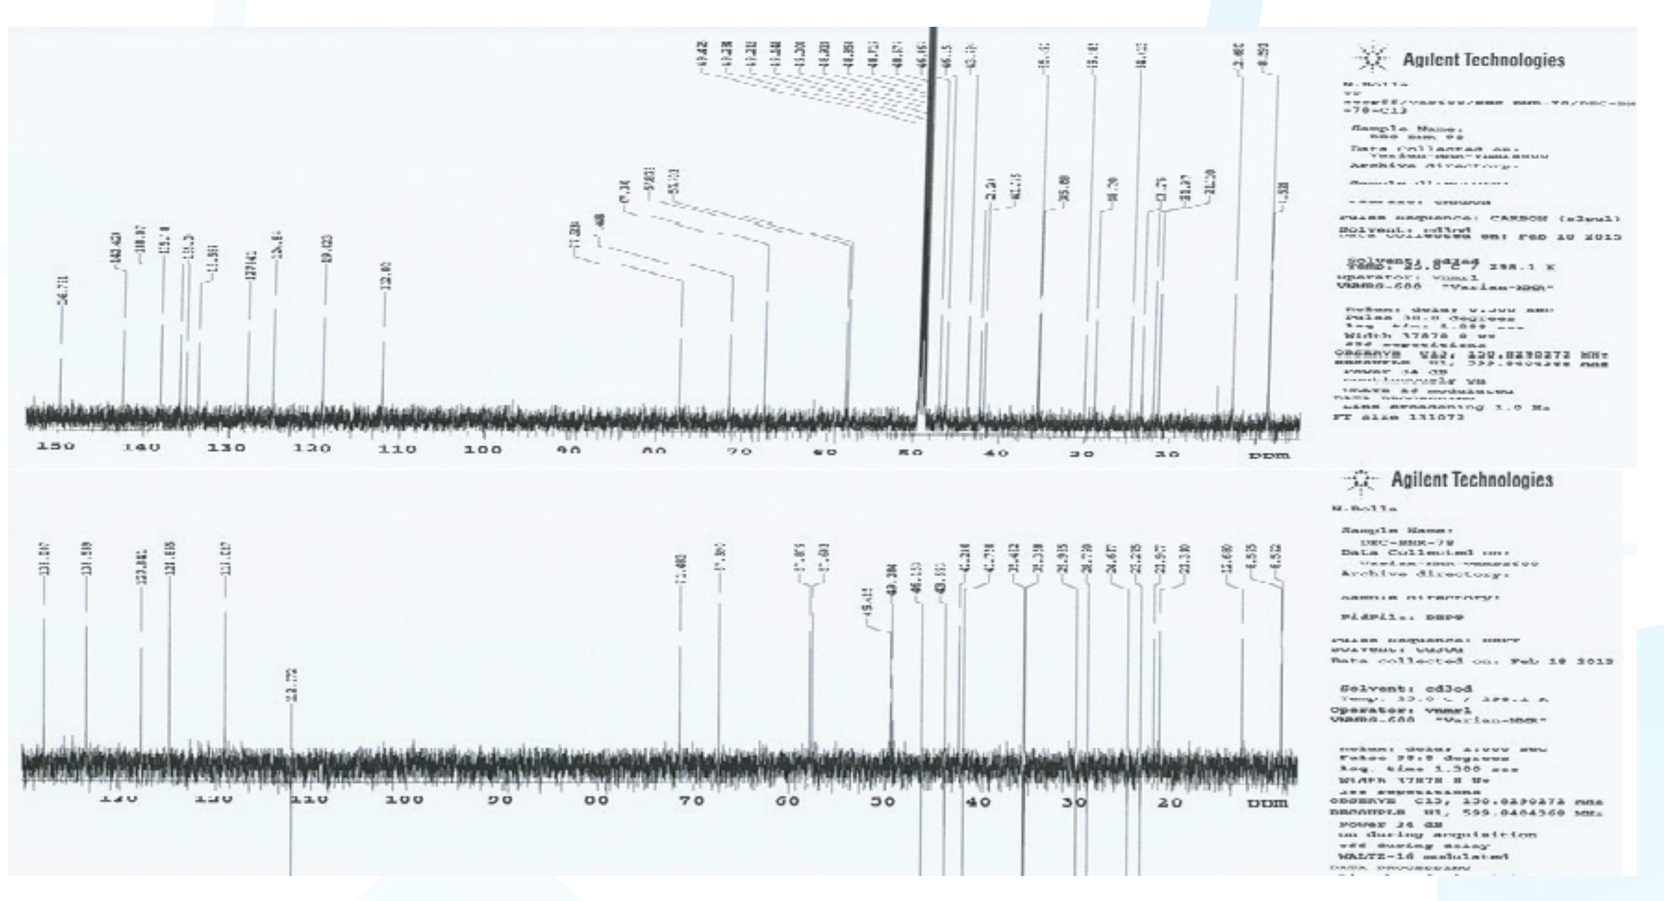

XI. MS and IR spectra of PRI-1917

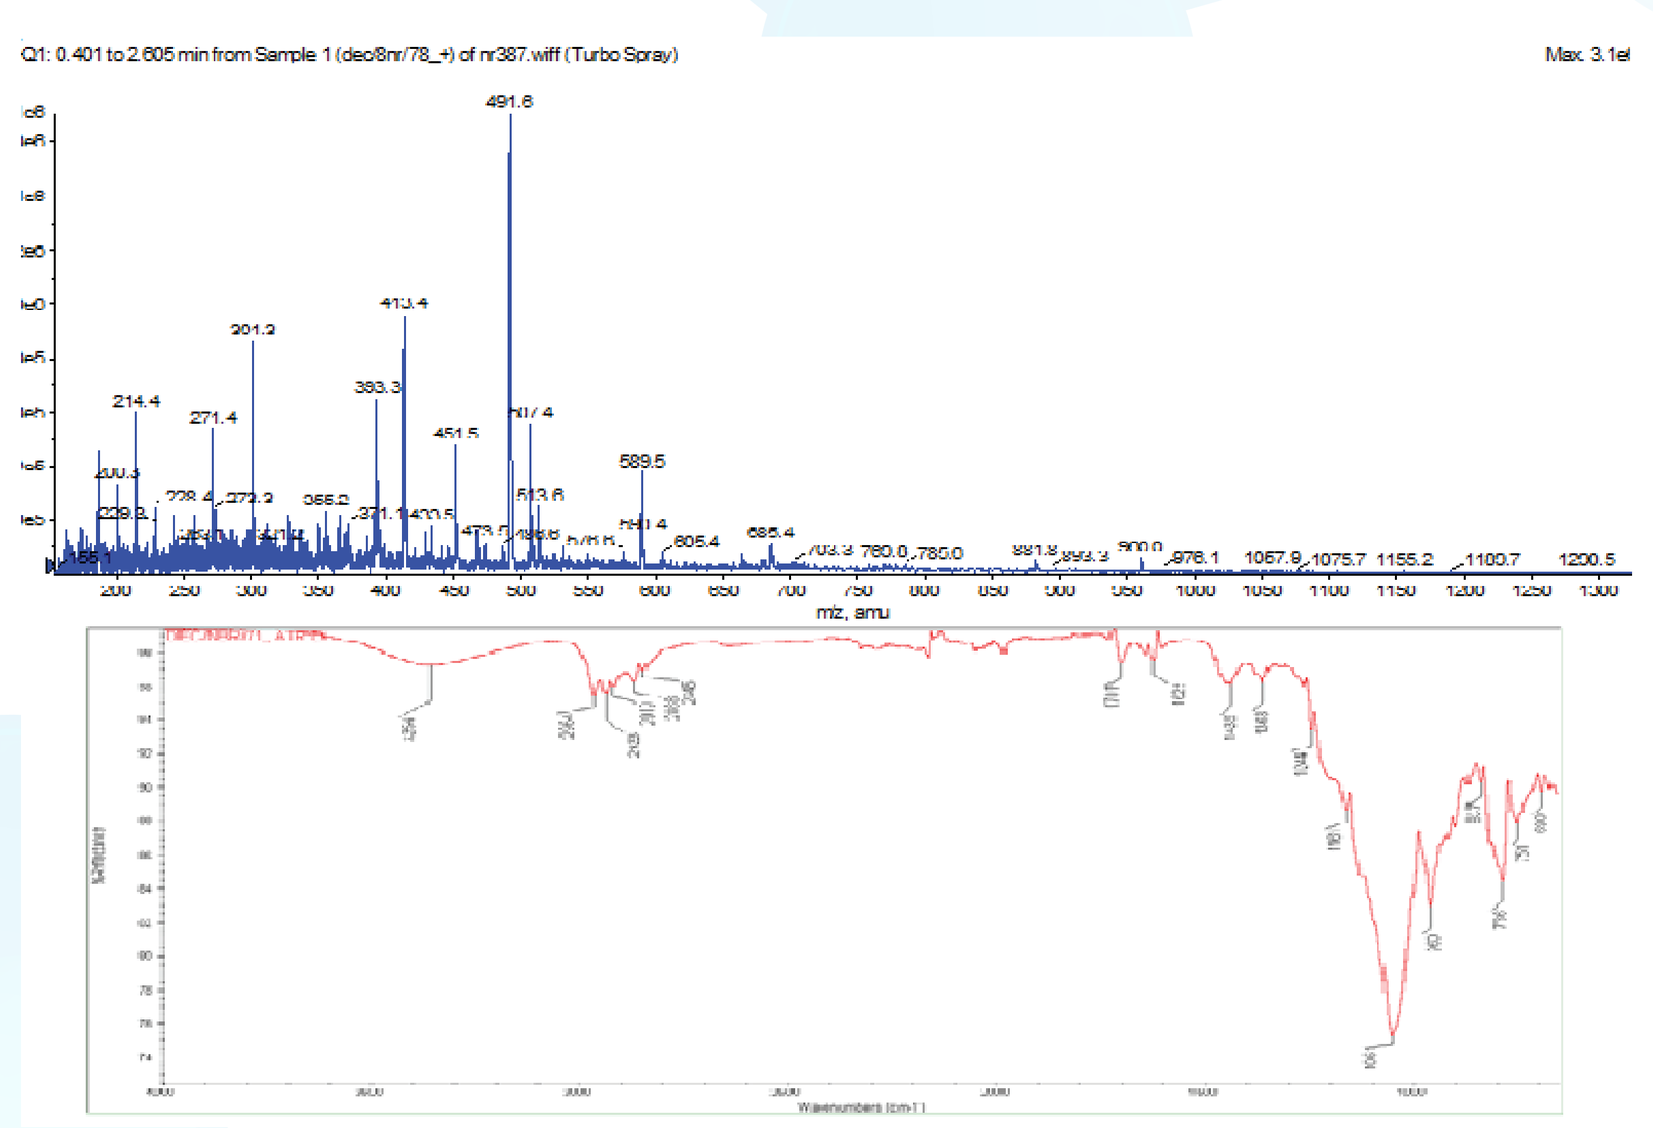

XII. VDR binding affinity test results:

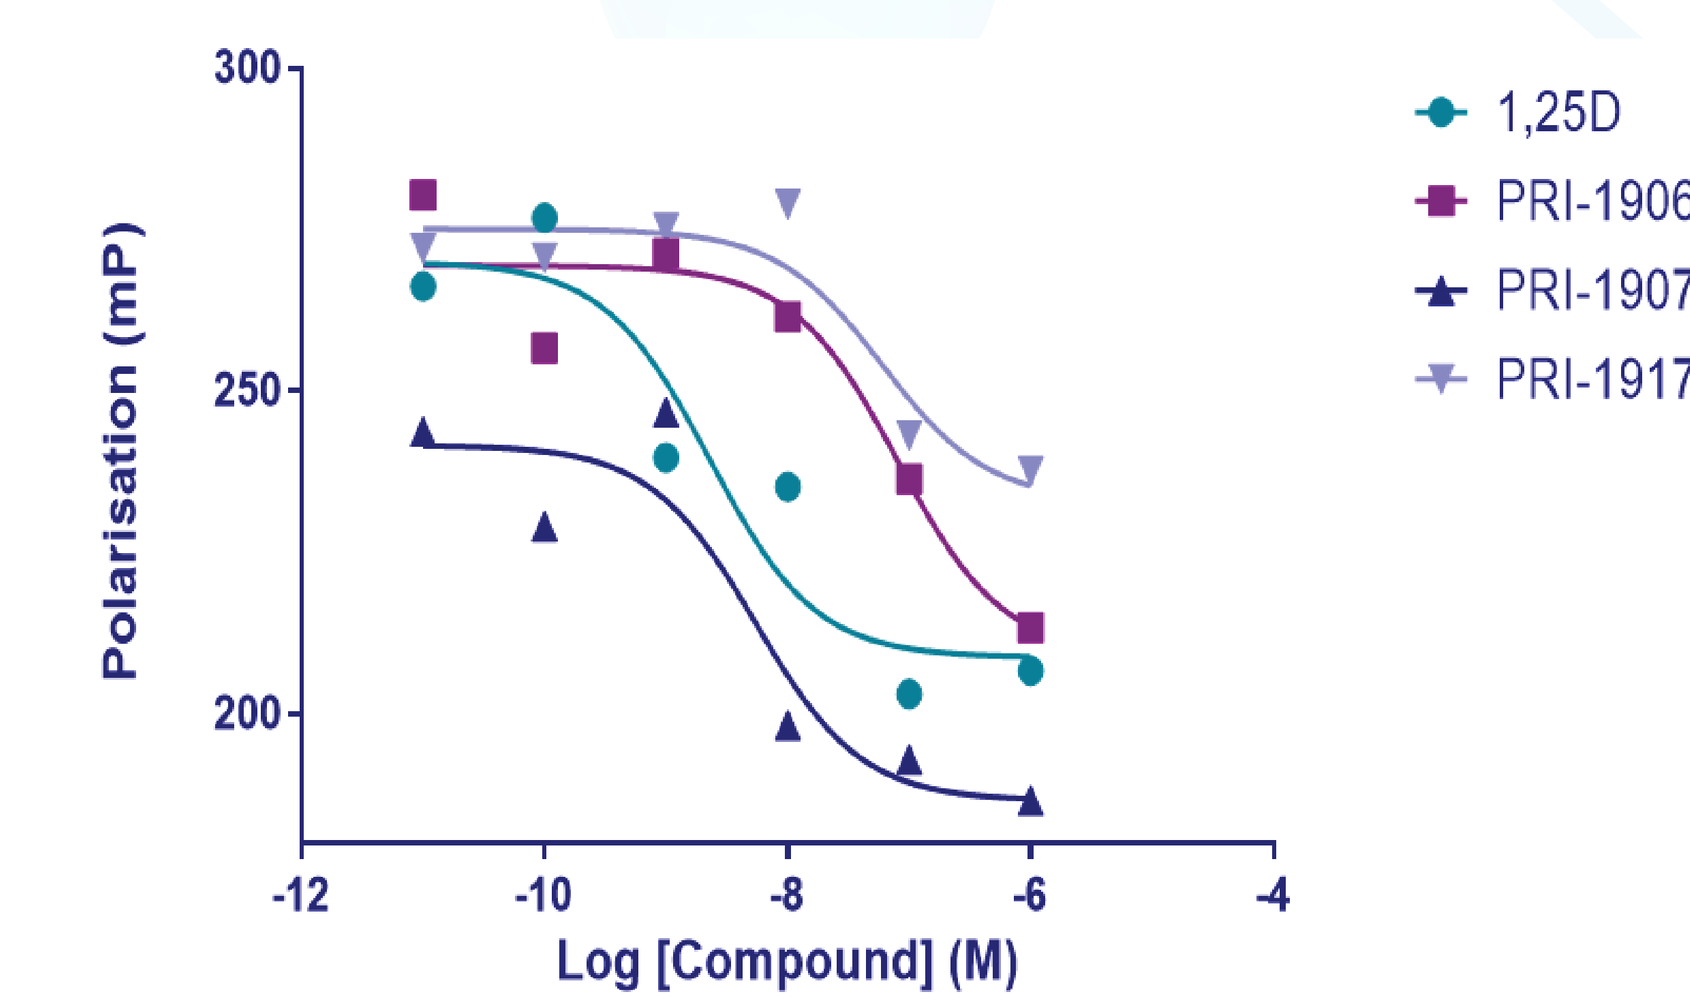

1,25D
PRI-1906
PRI-1907
PRI-1917
Polarisation (mP)
Log [Compound] (M)

	1,25 $(OH)_2D_3$	PRI-1906	PRI-1907	PRI-1917
IC_{50}	2.292e^{-09}	8.032e^{-08}	6.172e^{-09}	6.03 e^{-08}
Relative binding affinity[a]	100	2.85	37.10	3.80

[a]Potency of 1,25$(OH)_2D_3$ normalized to 100

XIII. Results and conclusions:

1. Two A-ring fragments were synthesized
2. Coupling conditions were optimized
3. PRI-1906 and PRI-1907 (potent vitamin D analogues) were synthesized in a new syn-thetic route
4. New 24a-geometricisomers of PRI-1906 and PRI-1907 were synthesized and characteri-sed with NOE NMR experiments, 1H NMR, 13C NMR, MS, HRMS, IR and UV spectroscopy.
5. VDR-binding affinity test results showed that 1,25$(OH)_2D_3$ has the strongest affinity to VDR, then 1907, followed by 1917 and finally 1906. So 1907 binds with higher affinity (the best) to VDR out of all three analogues. Order of binding affinity from higher to lower was 1,25$(OH)_2D_3$>1907>1917>1906.

XIV. References:

1. A.Kutner; 9th workshop on vitamin D, Orlando, Florida, USA(1994).
2. Bolla, N. R.; Marcinkowska, E.; Brown, G.; Kutner, A., Retiferols – synthesis and biologi cal activity of a conceptually novel class of vitamin D analogs. *Expert Opinion on Thera peutic Patents* **2014**, *24* (6), 633-646.
3. Plonska-Ocypa, K.; Sibilska, I.; Sicinski, R. R.; Sicinska, W.; Plum, L. A.; DeLuca, H. F., 13,13-Dimethyl-*des*-C,D analogues of (20*S*)-1α,25-dihydroxy-2-methylene-19-nor-vitamin D_3 (2MD): Total synthesis, docking to the VDR, and biological evaluation. *Bioorganic & Medi cinal Chemistry* **2011**, *19* (23), 7205-7220.

Acknowledgements:

The research leading to these results has received funding from the People Programme (Marie Curie Actions) of the European Union's Seventh Framework Programme FP7/2007--2013 under REA grant agreement number 315902. One author (NRB) gratefully acknowled-ges receipt of a Marie Curie Research Associate post. A Kutner, E Marcinkowska and G Brown are partners within the Marie Curie Initial Training Network DECIDE (Decision-making within cells and differentiation entity therapies).